Population Pharmacokinetic Modeling of Investigational Agent MLN4924 in Cancer Patients

H. Steve Kuan,[1] R. Donald Harvey[2]

[1]Department of Clinical Pharmacology, Millennium Pharmaceuticals, Inc., Cambridge, MA, USA; [2]Winship Cancer Institute of Emory University, Atlanta, GA, USA

INTRODUCTION

- MLN4924 is a novel investigational inhibitor of NEDD8-activating enzyme[1] that is being evaluated in patients with hematologic and non-hematologic cancer.

OBJECTIVE

- To characterize the pharmacokinetics (PK) of MLN4924 administered as a 1-hour intravenous infusion and to assess the effects of patient characteristics and assigned dose.

METHODS

Data collection

- Adult patients were treated for 21-day cycles in four ongoing dose-escalation clinical studies.
 - MLN4924 was infused intravenously on one of five dosing schedules (days 1–5; days 1, 3, 5; days 1, 2, 8, 9; days 1, 8; or days 1, 4, 8, 11).
- Serial or sparse blood samples were collected before and after the start of selected infusions during the first treatment cycle.
- MLN4924 concentrations were determined in plasma using a validated liquid chromatography with tandem mass spectrometry method.

Model development

- A population PK model was developed using NONMEM 7.1.2 with the FOCE interaction estimation method.[2]
- Two- and three-compartment disposition and zero-order input models were evaluated with or without log-transformation of concentration data.
- Drug concentrations below the lower limit of quantification (1 ng/mL) were treated as missing.
- Pre-specified covariates (Table 1) were incorporated in PK parameters as:
 - Power or linear-normalized models for continuous covariates, after centering on the median
 - Fractional models for categorical covariates.

Table 1. Pre-specified covariates of interest

Continuous variables	Median (range)
Age (AGE, years)	60.0 (29.3–90.8)
Body surface area (BSA, m²)	1.99 (1.48–2.72)
Body-mass index (BMI, kg/m²)	28.2 (17.9–45.6)
Body weight (WTKG, kg)	81.3 (51.4–147.9)
Alanine transaminase (ALT, U/L)	20 (6–134)
Aspartate transaminase (AST, U/L)	25 (10–110)
Bilirubin (BILI, μmol/L)	8.55 (1.71–18.8)
Creatinine clearance (CRCL, mL/min)	84.5 (5.47–150)
Assigned dose (TRTD, mg/m²)	67 (25–278)
Categorical variables	**N**
Sex (SEX)	M 67, F 39

- Effects of the covariates were investigated in stepwise procedures using likelihood ratio testing of nested models with one degree of freedom:
 - Forward addition (added if >3.84 decrease in the minimum value of the objective function)
 - Backward removal (removed if <10.83 increase in the minimum value of the objective function).

Model evaluation

- Goodness-of-fit plots were examined during the model-building process.
- Visual predictive checking (1000 simulations under the final model) was used for internal validation.

RESULTS

- One hundred and six (106) patients provided 1285 quantifiable concentrations, with a median of 11.5 values per patient (range 2–19).
- A two-compartment model parameterized by systemic clearance (CL), central volume of distribution (V_1), inter-compartmental clearance (Q), and peripheral volume of distribution (V_2) with log-transformed concentrations reasonably described the data (Figure 1).

Figure 1. Goodness-of-fit plots for the final model

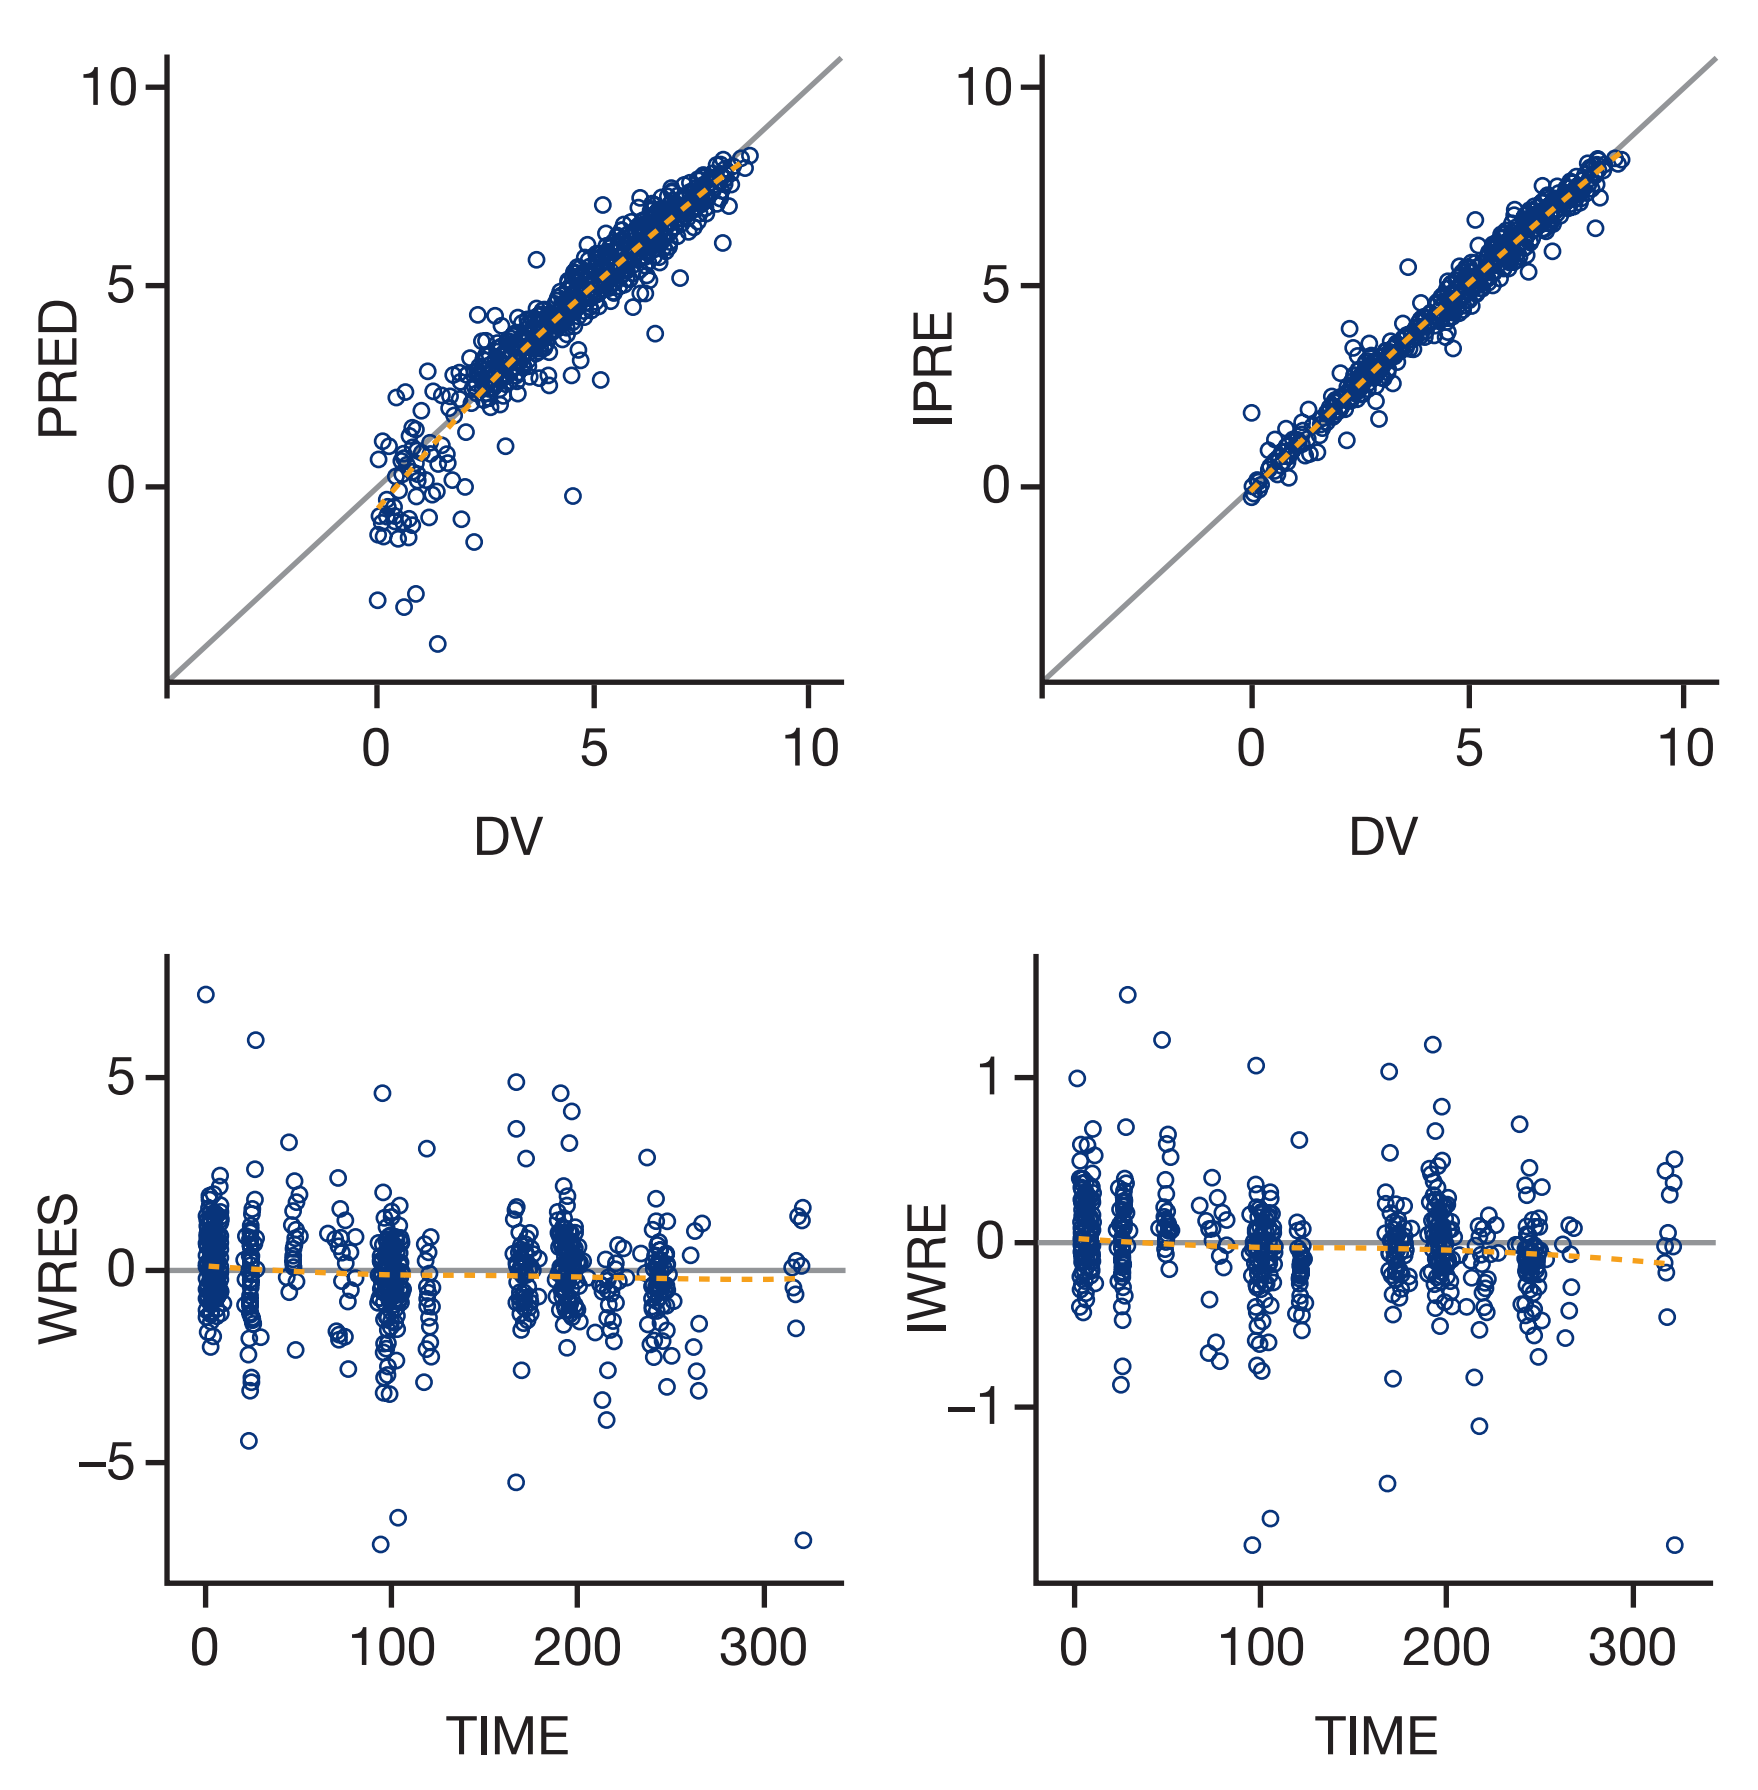
PRED
IPRE
DV
WRES
IWRE
TIME

- PK and variability parameters were estimated with reasonable precision in the base and final models (Table 2).

Table 2. Estimated population PK parameters of MLN4924

	Base model		Final model	
Parameter	Estimate	RSE (%)	Estimate	RSE (%)
CL (L/h)	42.2	3.5	59.6	8.2
V_1 (L)	132	6.8	139	6.0
Q (L/h)	35.5	8.7	35.2	8.9
V_2 (L)	161	4.9	187	5.3
IIV_{CL}	33.8%	21.4	26.7%	23.2
IIV_{V1}	61.0%	20.5	42.9%	31.7
IIV_Q	50.5%	34.4	53.2%	30.2
IIV_{V2}	35.9%	26.9	27.6%	35.3
RV	0.277	8.7	0.275	8.7

IIV=inter-individual variability reported as CV; RSE=percent relative standard error; RV=residual variability (additive).

- AGE, TRTD, BSA, and SEX were identified as statistically significant covariates (Table 3).

Table 3. Significant relationships of MLN4924 PK parameters with covariates

Parameter	Covariate	Estimate	RSE (%)
CL	BSA	0.948	16.5
	AGE	–17.3	26.9
V_1	BSA	2.37	14.4
	TRTD	–0.385	21.2
V_2	SEX	–0.374	11.7

$CL = 59.6 \cdot \left(\frac{BSA}{1.99}\right)^{0.948} - 17.3 \cdot \left(\frac{AGE}{60}\right)$ $\quad V_1 = 139 \cdot \left(\frac{BSA}{1.99}\right)^{2.37} \cdot \left(\frac{TRTD}{67}\right)^{-0.385}$

$V_2 = 187 \cdot (1 - 0.374 \cdot SEX\ [0 \text{ if male; } 1 \text{ if female}])$.

- CL decreased with increasing AGE and increased with increasing BSA, with both covariates explaining 7.1% of its inter-individual variability (Figure 2).

Figure 2. Relationships of MLN4924 systemic clearance with body surface area and age

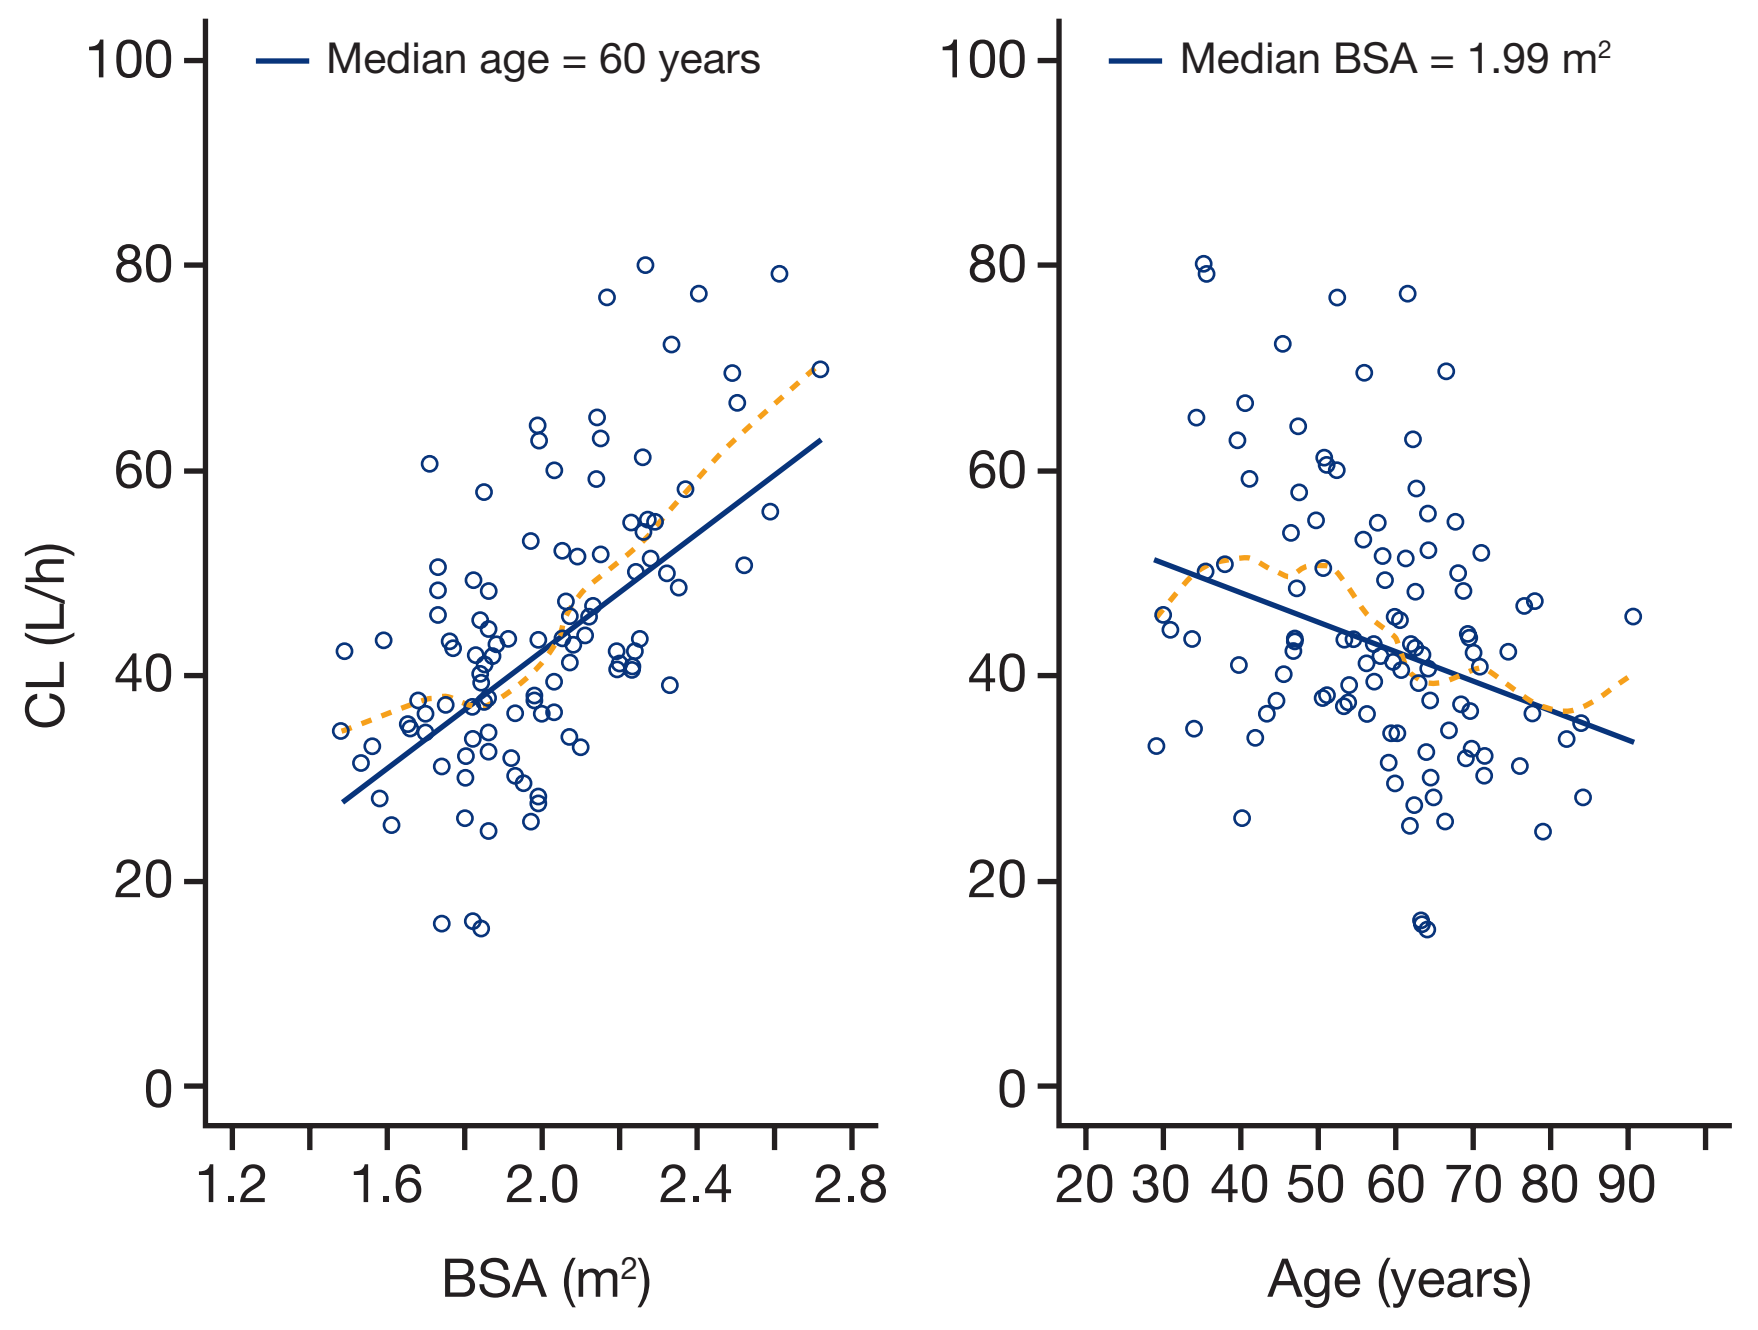
Median age = 60 years
Median BSA = 1.99 m²
CL (L/h)
BSA (m²)
Age (years)

- V_1 decreased with increasing TRTD and increased with increasing BSA, with both covariates explaining 18.1% of its inter-individual variability (Figure 3).

Figure 3. Relationships of MLN4924 central volume of distribution with BSA and assigned dose

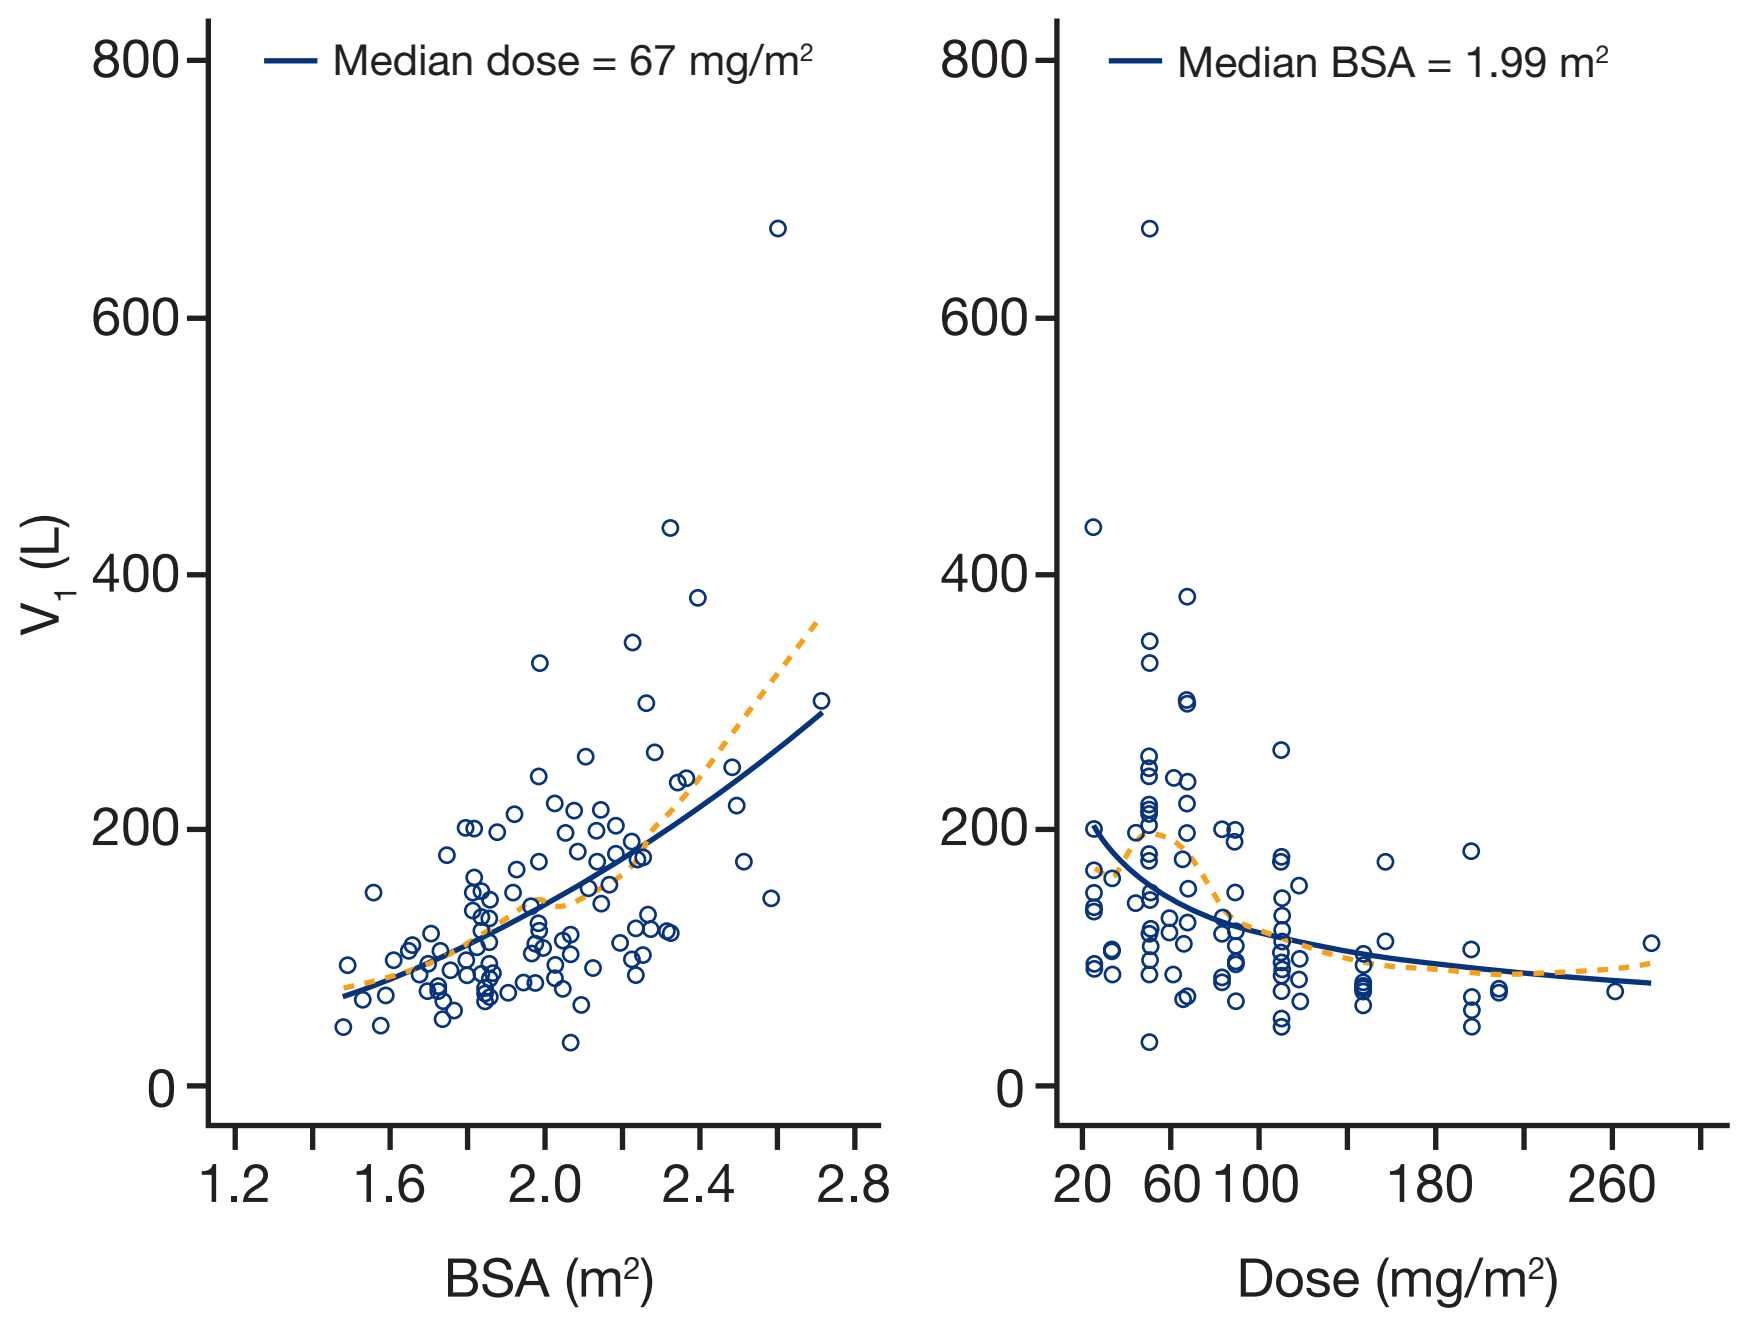
Median dose = 67 mg/m²
Median BSA = 1.99 m²
V1 (L)
BSA (m²)
Dose (mg/m²)

- V_2 was higher for males than for females, with SEX explaining 8.3% of its inter-individual variability (Figure 4).

Figure 4. Difference in peripheral volume of distribution of MLN4924 between sexes

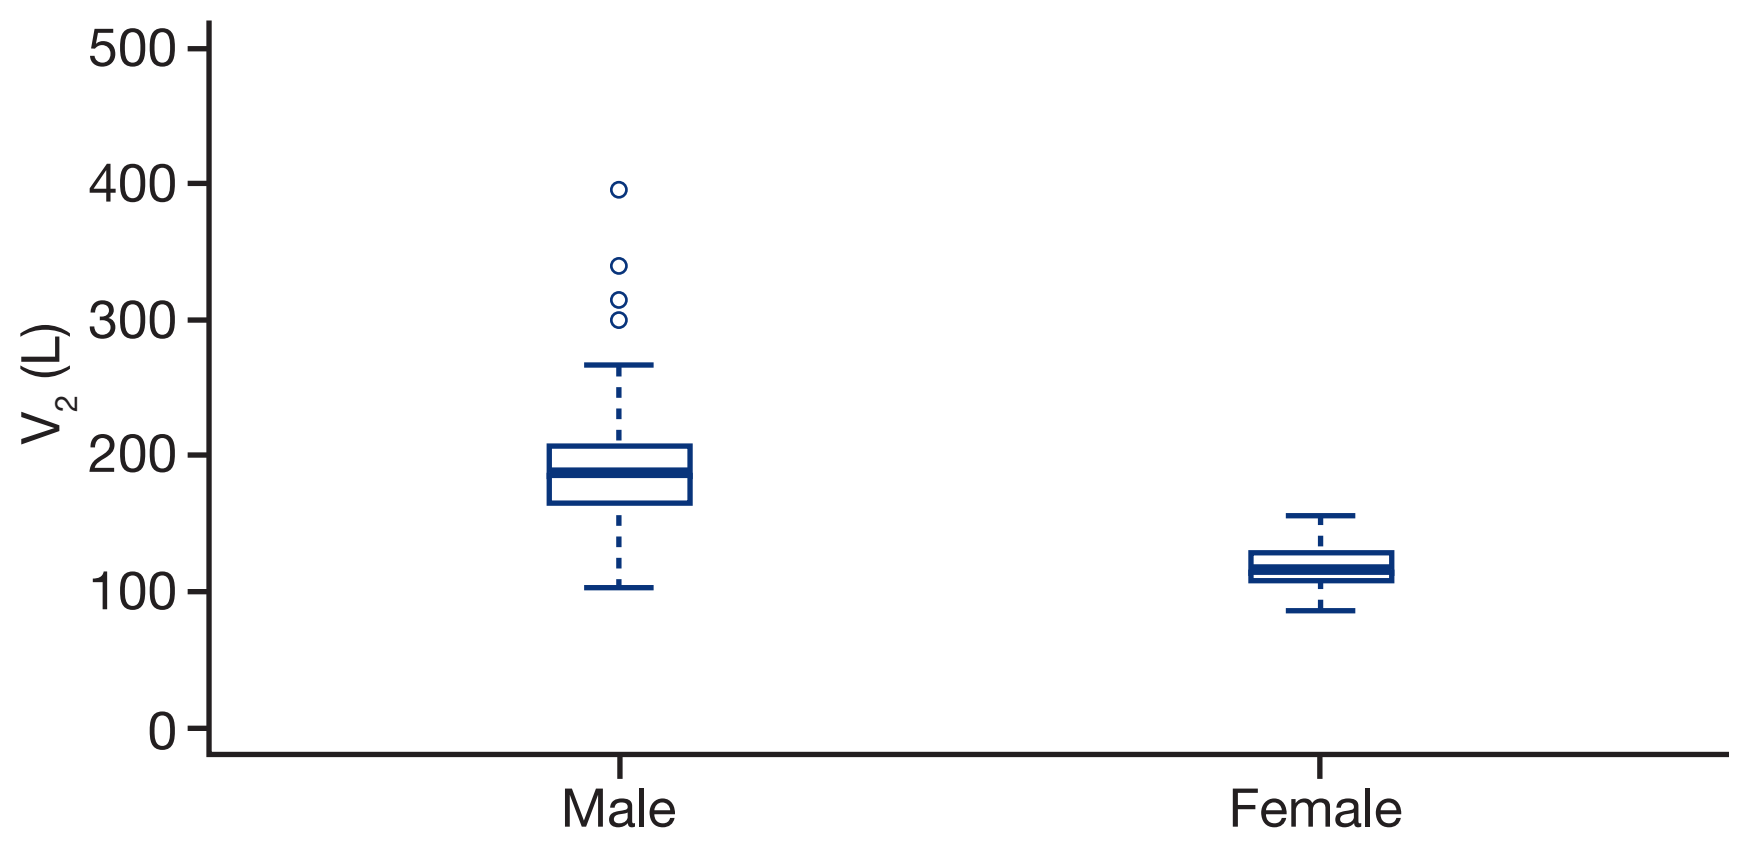
V2 (L)
Male
Female

- Predictive performance of the final model was reasonable, as the 90% prediction interval contained the 5th and 95th percentiles of the observed data (Figure 5).

Figure 5. Simulated and observed MLN4924 concentrations versus time

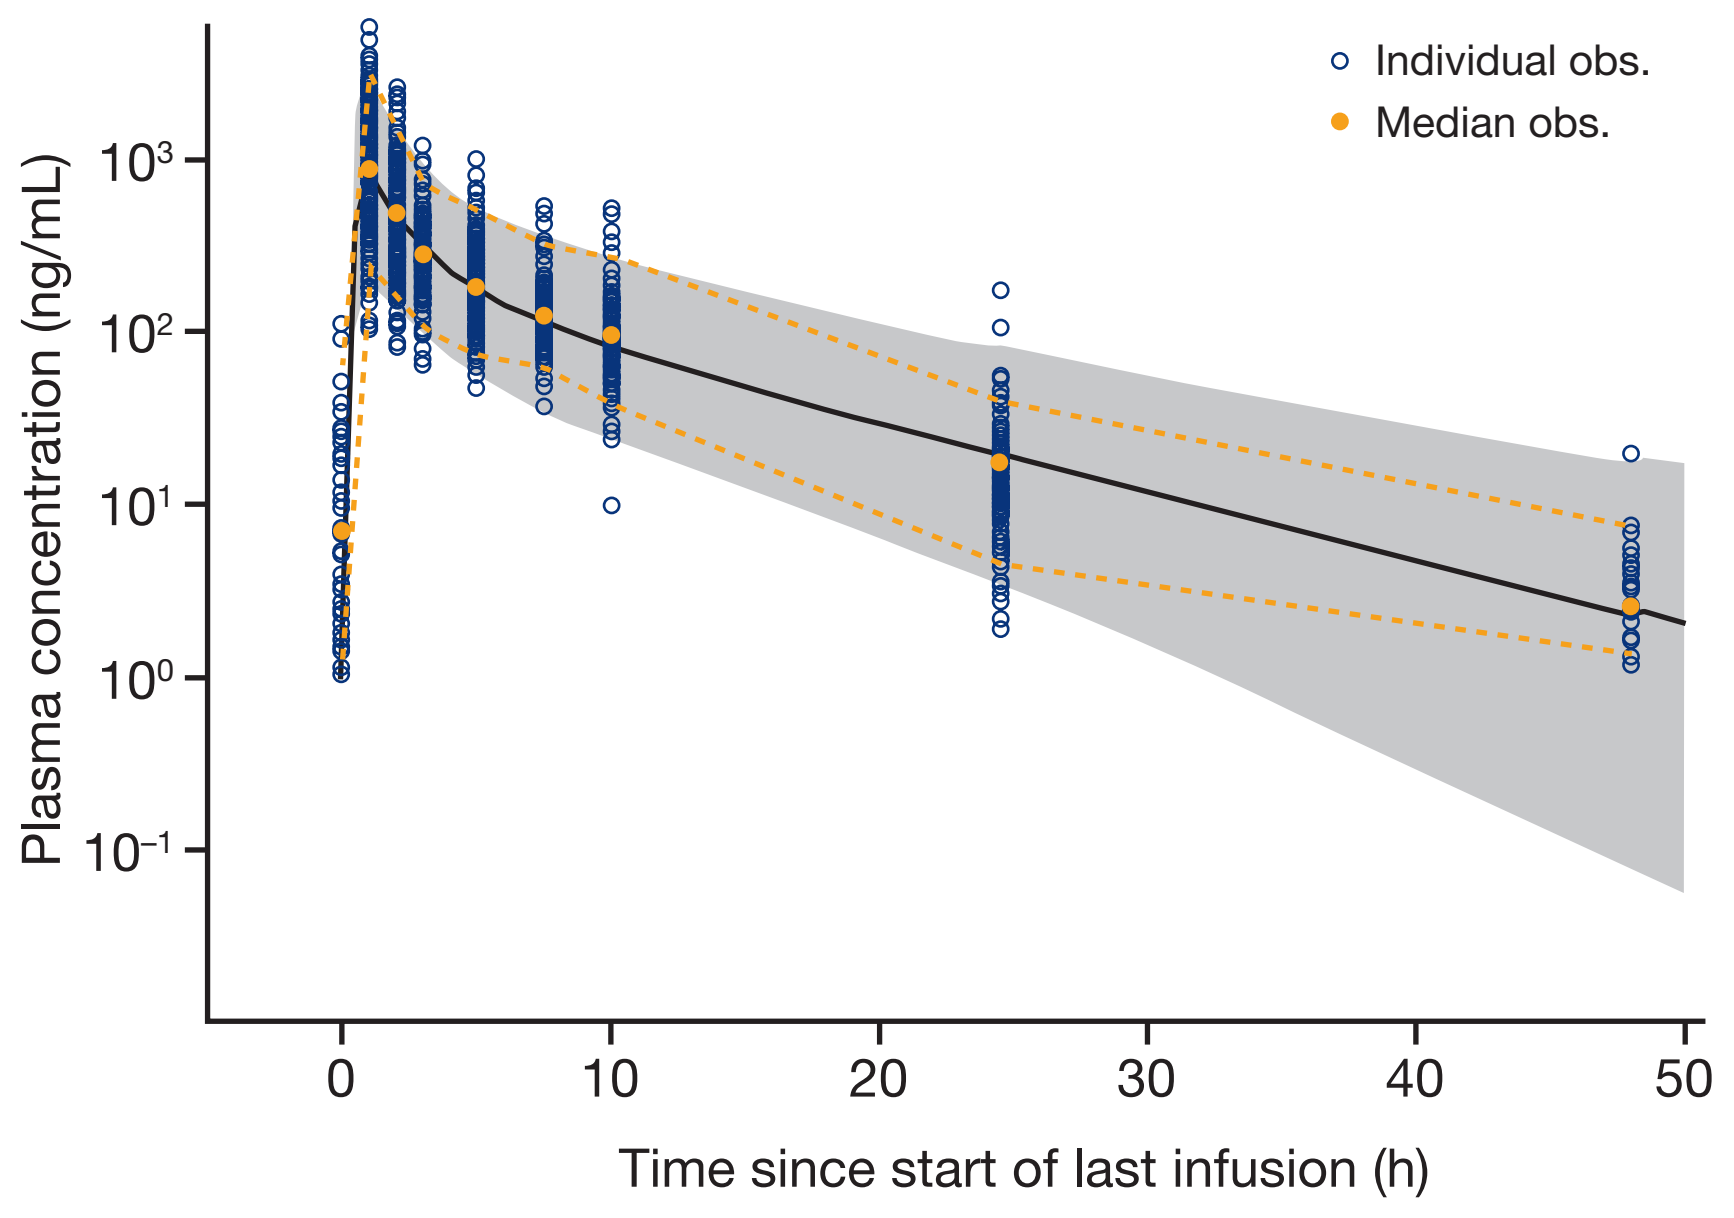
Individual obs.
Median obs.
Plasma concentration (ng/mL)
Time since start of last infusion (h)

The black line and gray area are median prediction and 95% prediction interval, respectively. The orange dashed lines represent the 5th and 95th percentiles of observed data.

CONCLUSIONS

- The effect of BSA was significant on both CL and V_1, lending support to MLN4924 administration on the basis of body size.
- The effect of age was significant on CL, resulting in higher plasma exposure to MLN4924 in the elderly.
- The effect of dose was significant on V_1, likely attributable to saturable binding in whole blood.
- The effect of SEX was significant on V_2 and the difference may not be explained by body size.
- Clinical relevance of the significant covariates will be further evaluated in the context of safety and efficacy.

REFERENCES

1. Milhollen MA, et al. MLN4924, a NEDD8-activating enzyme inhibitor, is active in diffuse large B-cell lymphoma models: rationale for treatment of NF-kappaB-dependent lymphoma. Blood 2010;116(9):1515-23.
2. Beal S, et al. NONMEM User's Guides. (1989–2009), Icon Development Solutions, Ellicott City, MD, USA, 2009.

ACKNOWLEDGMENTS

The authors acknowledge the editorial assistance of Steve Hill of FireKite in the development of this poster, which was supported by Millennium Pharmaceuticals, Inc.

Poster presented at the Population Approach Group in Europe (PAGE) meeting, Athens, Greece, 7–10 June, 2011.

23386F Kuan PAGE Poster_v3.indd 1 28/06/2011 14:21